Plain Language Summary of ENGOT-EN6/GOG-3031/ NSGO-RUBY: A Phase 3, Randomized, Double-Blind, Multicenter Study of Dostarlimab + Carboplatin–Paclitaxel Versus Placebo + Carboplatin–Paclitaxel in Recurrent or Primary Advanced Endometrial Cancer

Table of Contents

- General information about this study
- Which patients can enroll?
- Why is this study being done?
- How will the trial be conducted?
- What medicine is being tested?
- Where can I find more information?

General information about this study

Full scientific title:
A Study of Dostarlimab (TSR-042) Plus Carboplatin-Paclitaxel Versus Placebo Plus Carboplatin-Paclitaxel in Patients With Recurrent or Primary Advanced Endometrial Cancer (RUBY)

Study number:	Study name:	Sponsored by:
NCT03981796	RUBY	GlaxoSmithKline

Writing and editorial support for this report was provided by Ashfield Healthcare Communications and funded by GlaxoSmithKline

Why is this study being done?

The RUBY study is being done to see if treatment with dostarlimab plus chemotherapy followed by dostarlimab alone is useful in treating endometrial cancer

What medicine is being tested?

Dostarlimab is a medicine being tested that is part of a category called programmed cell death inhibitors (or "PD-1 inhibitors"). These drugs help your immune system to find and attack cancer cells

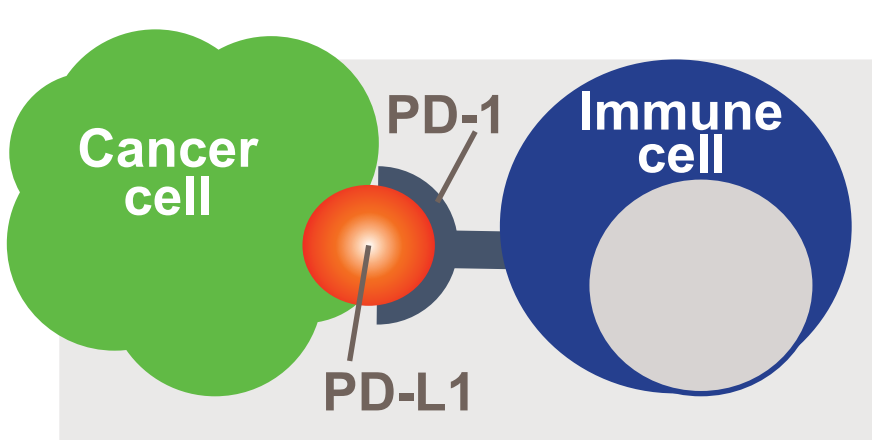

Cancer cell
PD-1
Immune cell
PD-L1

When cancer cells have PD-L1 on their cell surface, immune cells that have PD-1 will find them, and they connect. Through this "handshake," the immune system is told not to attack the cancer cells

Dostarlimab attaches to PD-1 and prevents the handshake. This immune cell can now signal to the rest of the immune cells that the cancer cells should be attacked

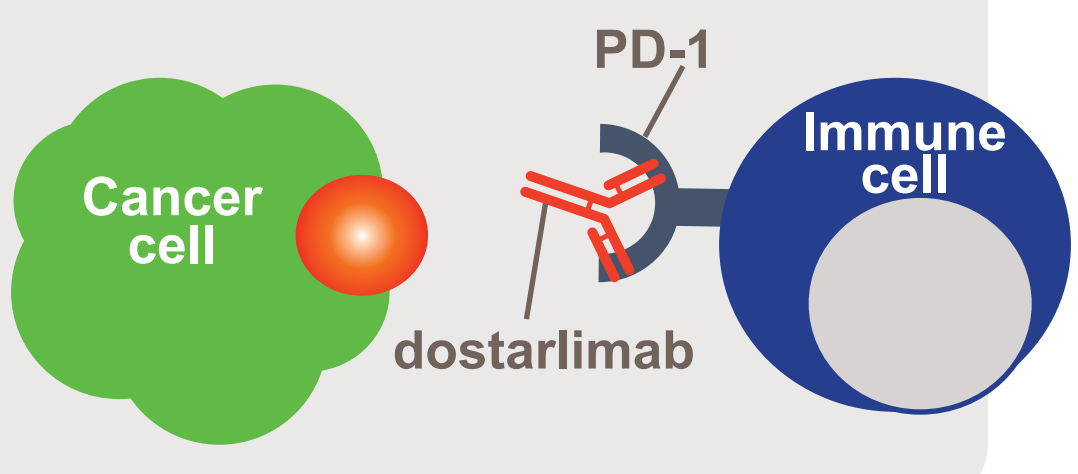

PD-1
Cancer cell
Immune cell
dostarlimab

Which patients can enroll?

Patients must have endometrial cancer that is advanced or recurrent

Advanced endometrial cancer means it is stage 3 or 4 disease when the cancer is first diagnosed

Stage 3

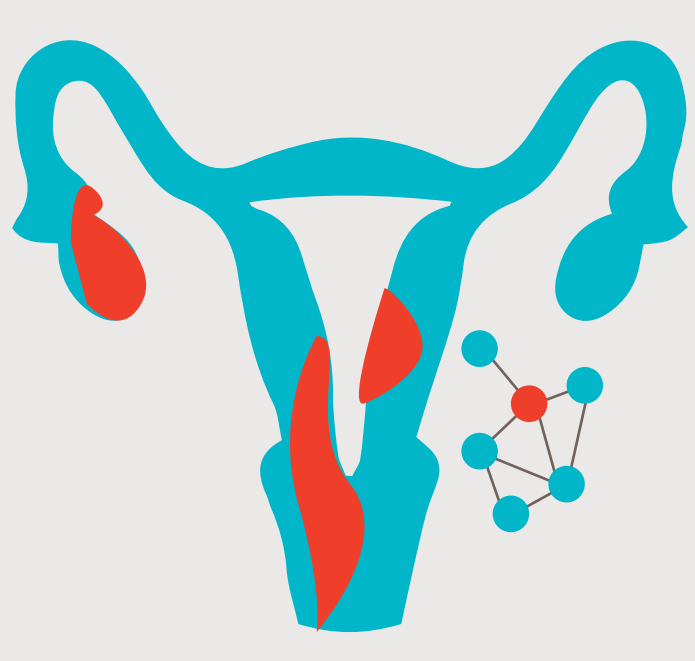

The cancer has spread to the ovary, vagina, or pelvic lymph nodes

Stage 4

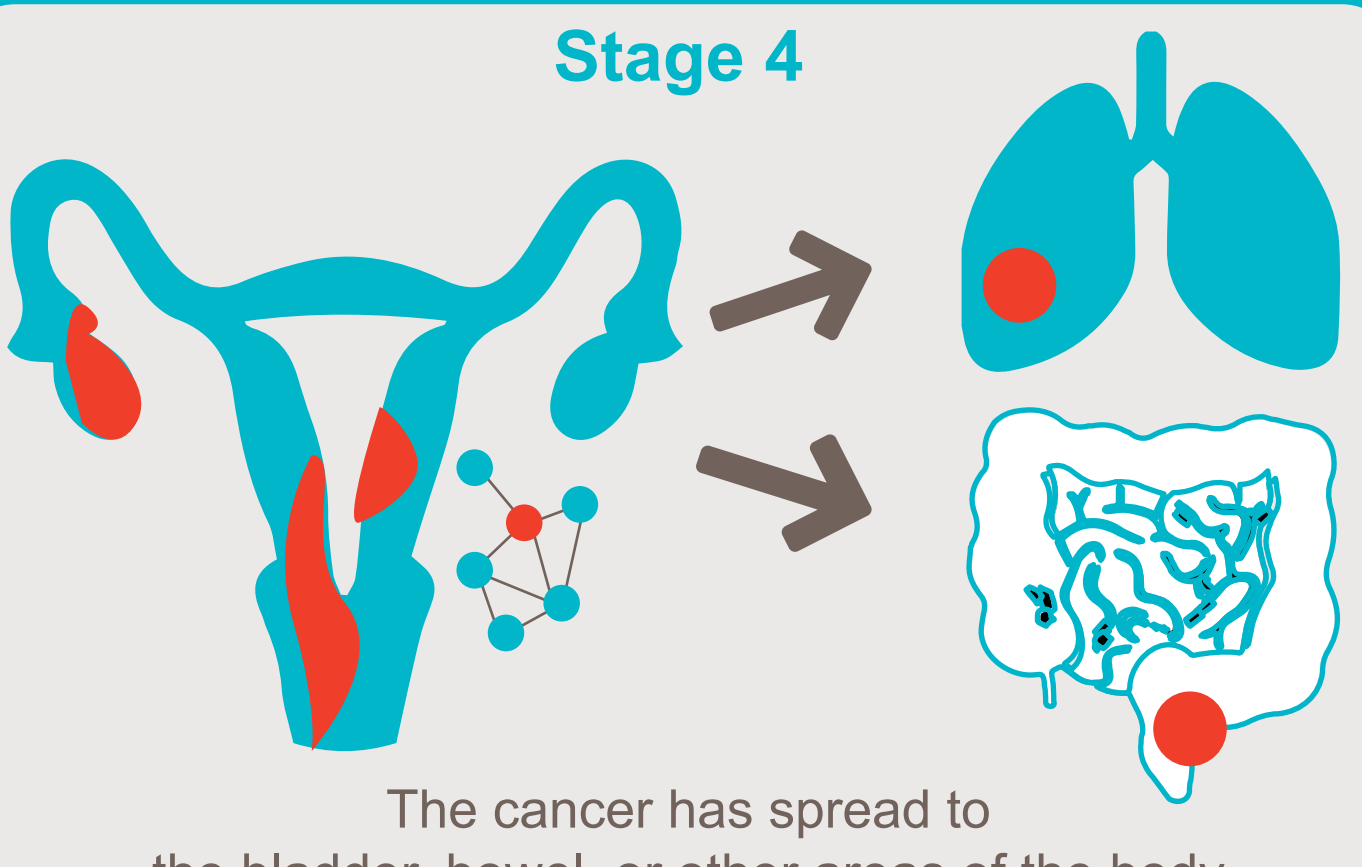

The cancer has spread to the bladder, bowel, or other areas of the body

Recurrent endometrial cancer means it was treated and has returned

Patients must be in good enough condition to take care of themselves on their own. For example, they must be able to walk or do light housework or office work

Not all patients who meet the criteria shown here will be eligible. More detailed information on who is eligible to participate is available at https://clinicaltrials.gov/ct2/show/NCT03981796

How will the trial be conducted?

Patients with recurrent or primary advanced endometrial cancer

Randomized 1:1

treatment option 1: Dostarlimab + carboplatin + paclitaxel → Dostarlimab

treatment option 2: Placebo + carboplatin + paclitaxel → Placebo

every 3 weeks, 6 cycles

every 6 weeks, up to 3 years

Analysis of results

RUBY is a randomized, double-blind, phase 3 trial. This means that there are **2 treatment options**

1. Dostarlimab plus chemotherapy (the treatment being tested)

2. Standard chemotherapy (the control)

- **Chemotherapy** is a drug treatment that uses powerful chemicals to kill cancer cells in your body
- There are **two chemotherapy drugs** that are used together in this trial: one is called **carboplatin**, and the other is called **paclitaxel**

- Patients will be **placed into the arms randomly** (this is what is meant by **"randomized"**)
- In the **control arm**, patients will receive a **placebo** (inactive treatment that looks the same as the dostarlimab treatment) so that neither the patient nor the oncologist know which treatment option the patient is getting (this is what is meant by **"double-blind"**)
- **Phase 3 trials** compare a new potential treatment against the current standard of care to see if the new treatment is **better than the current standard of care**
- More details on clinical trial phases can be found here
- **The trial is enrolling at sites in several countries**

Belarus	Greece	Sweden
Belgium	Hungary	Turkey
Canada	Israel	Ukraine
Czech Republic	Italy	United Kingdom
Denmark	Netherlands	United States
Finland	Norway	
Germany	Poland	

Where can I find more information?

https://clinicaltrials.gov/ct2/show/NCT03981796